A cocktail of two heavy chain-only antibodies targeting 3 epitopes spread across the spike protein S1 and S2 subunits that neutralizes all SARS-CoV-2 variants tested so far with exceptional potency

Rinaldi M.[1], Venneman T.[1], Van Royen T.[1], Blanchetot C.[1], Herschke F.[1], Schepens B.[2,3], De Cae S.[2,3], Rossey I.[2,3], Roose K.[2,3], van Schie L.[2,3], Reiter D.[4,5], Remaut H.[4,5], Callewaert N.[2,3], Saelens X.[2,3] and Bockstal V.[1]

1 ExeVir Bio BV, Ghent, Belgium
2 VIB-UGent Center for Medical Biotechnology, Ghent, Belgium
3 Department of Biochemistry and Microbiology, Ghent University, Ghent, Belgium
4 Structural Biology Brussels, Vrije Universiteit Brussels, Belgium
5 VIB-VUB Center for Structural Biology, Brussels, Belgium

Funded by the European Union

Funding:
This project received funding from the EU's Horizon 2021 research and innovation programme (HORIZON-HLTH-2021-CORONA-01), grant agreement No 101045949

Introduction: More than 7 million people have died from COVID-19 to date and SARS-CoV-2 continues to cause substantial disease around the world. High-risk patients like immunocompromised and elderly are often not able to elicit adequate immune responses to vaccination and strongly benefit from an additional layer of protection in the form of complementary antibody therapy. Monoclonal antibodies act immediately and can provide immune support for months. However, all previously authorized antibodies lost their neutralization potency against the currently circulating and constantly mutating SARS-CoV-2 variants, leaving a vast unmet medical need.
M&M: The combination XVR012 consists of the molecules XVR013m and XVR014. XVR013m is a mono-specific VHH-Fc antibody that targets a unique epitope in the S2 subunit, XVR014 is a bi-specific bivalent VHHx-Fc-VHHy antibody construct, targeting 2 non-competing epitopes in the Receptor-Binding Domain of the spike. Both antibodies contain a human IgG1 Fc with LS mutations for half-life extension. XVR012 delivers a triple mode of action in preventing infection of host cells, i.e., (1) sterical hinderance of ACE2 receptor binding, (2) induction of S1 subunit shedding thereby preventing viral attachment and (3) inhibition of the fusion process between the viral and host cell membrane.
Results: XVR012 broadly neutralizes SARS-CoV-1 and SARS-CoV-2 viruses in vitro and demonstrates an IC50 ranging from 4.8 to 8,7 ng/mL against all SARS-CoV-2 variants tested so far in a pseudovirus neutralization assay, including the currently circulating BA.2.86.1, HK.3, EG.5.1 and HV.1. In vivo efficacy of XVR012 is demonstrated in a Syrian Golden Hamster challenge model in both prophylactic and therapeutic settings. PK parameters have been determined in a Tg32 SCID mouse model and support an envisioned duration of protection up to 6 months.
Conclusion: This second-generation cocktail targeting three unique and highly conserved epitopes in the S1 and S2 subunits of the spike is ready to proceed to clinical testing and may provide a long-term solution to the populations at highest risk.

1. Combination XVR012 consists of the molecules XVR013m and XVR014

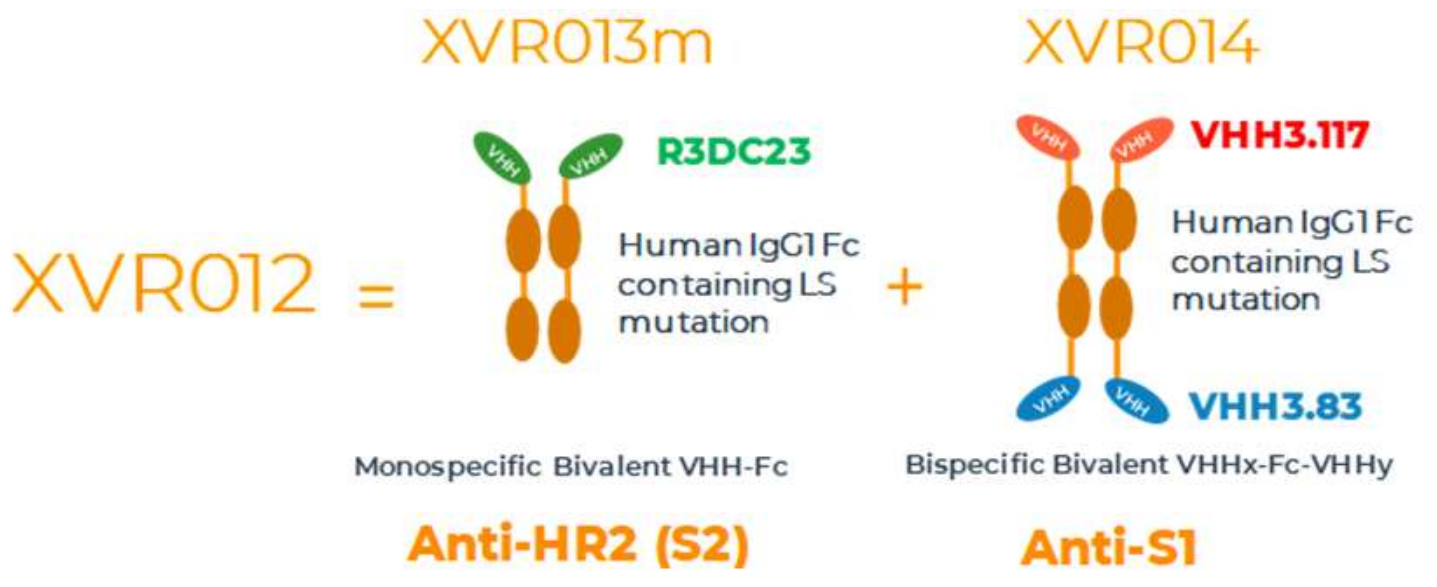
XVR012 =
XVR013m
R3DC23
Human IgG1 Fc containing LS mutation
+
XVR014
VHH3.117
Human IgG1 Fc containing LS mutation
VHH3.83
Monospecific Bivalent VHH-Fc
Bispecific Bivalent VHHx-Fc-VHHy
Anti-HR2 (S2)
Anti-S1

XVR012 is a potential best-in-class long-acting ultrapotent combination cocktail of **XVR013m**, a heavy chain-only VHH-Fc molecule targeting the highly conserved HR2 region of the S2 subunit, and **XVR014**, a bispecific heavy-chain only VHHx-Fc-VHHy antibody targeting 2 non-competing epitopes in the RBD (S1 subunit).

2. Structural analysis of the binding to the spike protein

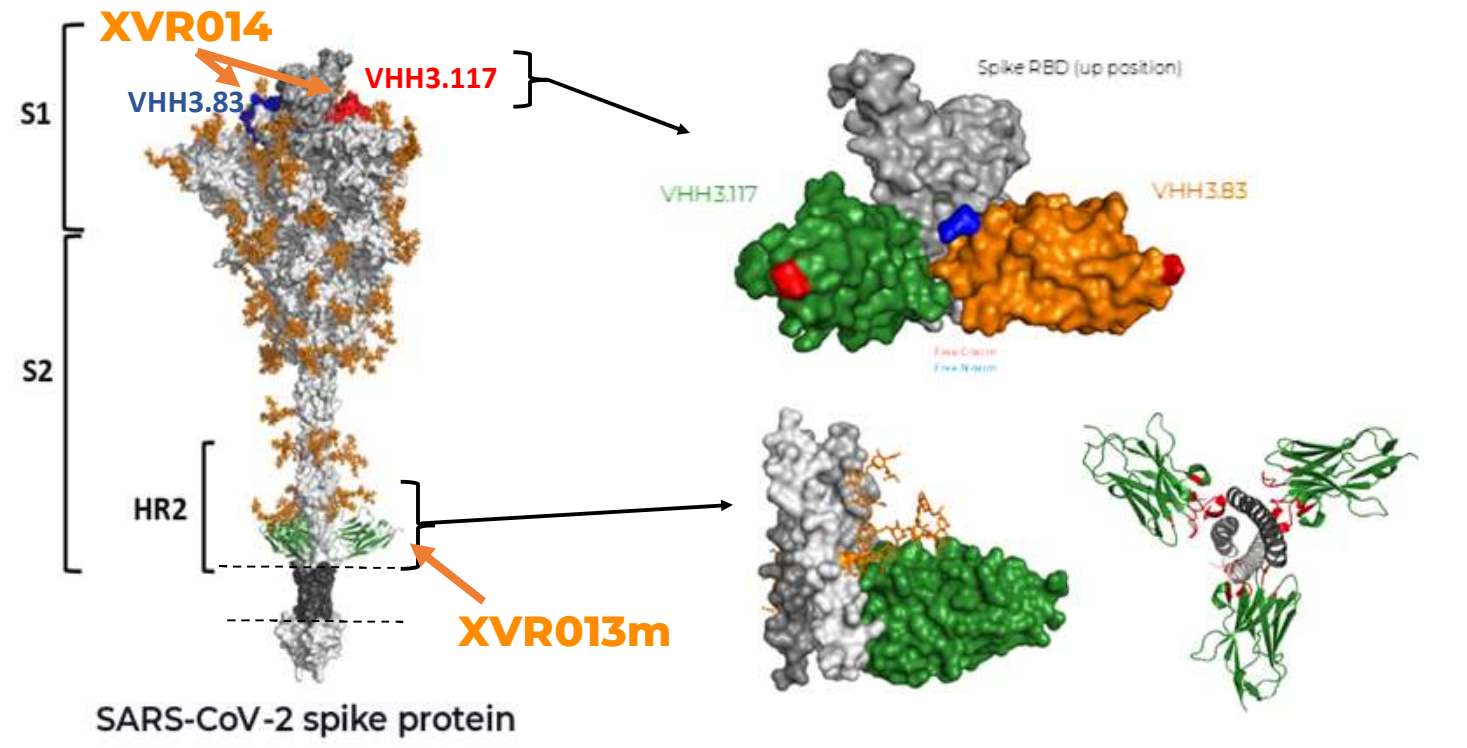
XVR014
VHH3.83
VHH3.117
S1
S2
HR2
XVR013m
SARS-CoV-2 spike protein
Spike RBD (up position)
VHH3.117
VHH3.83

VHH R3DC23 bound to a quaternary epitope in the HR2 domain of the S2 subunit of the spike protein between the terminal N11994 glycan (orange) and the viral membrane (dark grey, indicated by dotted lines), as determined by co-crystal structure (*De Cae et. al. BioRxiv 2023*). VHH3.117 and VHH3.83 bound to one RBD of the spike protein, as determined by cryo-EM.

3. Exceptional virus neutralization potency against all variants tested

Variant	Mean IC50 (ng/mL)	XVR012 =XVR013m+XVR014	XVR013m	XVR014
Reference strain	D614G	12,2	6,4	57,3
Omicron	XBB.1.5	7,2	3,6	87,6
	XBB.1.9.1	7,2	3,6	87,6
	XBB.1.9.2	7,2	3,6	87,6
	XBB.2.3	7,7	4,0	75,6
	XBB.1.16	7,1	3,6	90,4
F456L mutation containing variants	EG.5.1	7,8	4,3	67,9
	HK.3	8,7	4,6	191,0
	HV.1	7,6*	3,0	64,7
	BA.2.74	6,1	3,5	42,8
	BA.2.86.1	4,8	3,0	11,6

Data are generated in VSV-based pseudovirus neutralization assay and presented as mean IC50 based on 3 independent experiments, except for (*) 1 independent experiment.

XVR012 (tested in a **1:1 ratio** of XVR013m and XVR014), as well as the individual components XVR013m and XVR014, demonstrate **high resilience against all SARS-CoV-2 variants tested**.

4. *In vivo* pharmacokinetics study in a Tg32 (hFcRn) SCID mouse model

XVR014	XVR013m
CL= 0,203 mL/h/kg V1= 50,2 mL/kg Q= 10,2 mL/h/kg V2= 70,1 mL/kg	CL= 0,558 mL/h/kg V1= 61,4 mL/kg Q= 10,1 mL/h/kg V2= 118 mL/kg
$t_{1/2}$= 16,7 – 18,3 days	$t_{1/2}$= 9,3 days

In the XVR014 study, six animals were included per group and two dose levels (2 mg/kg and 10 mg/kg) were tested using a single intravenous (IV) administration. In the XVR013m study , 5 animals were included per group and a 10 mg/kg dose level was tested using a single IV administration. A two-compartment population PK model with linear elimination was used to model the PK parameters, including random effect terms on the elimination clearance (CL), central and peripheral volume of distribution (V1 and V2, respectively), and distribution clearance Q. Model evaluation was based on model diagnostics and goodness-of-fit plots. Allometric scaling was then applied to project the terminal half-life in human.

Predictions in human:
XVR013m $t_{1/2}$= 44-47 days
XVR014 $t_{1/2}$= 73-77 days

4. *In vivo* efficacy demonstrated in Syrian Golden Hamster SARS-CoV-2 challenge model

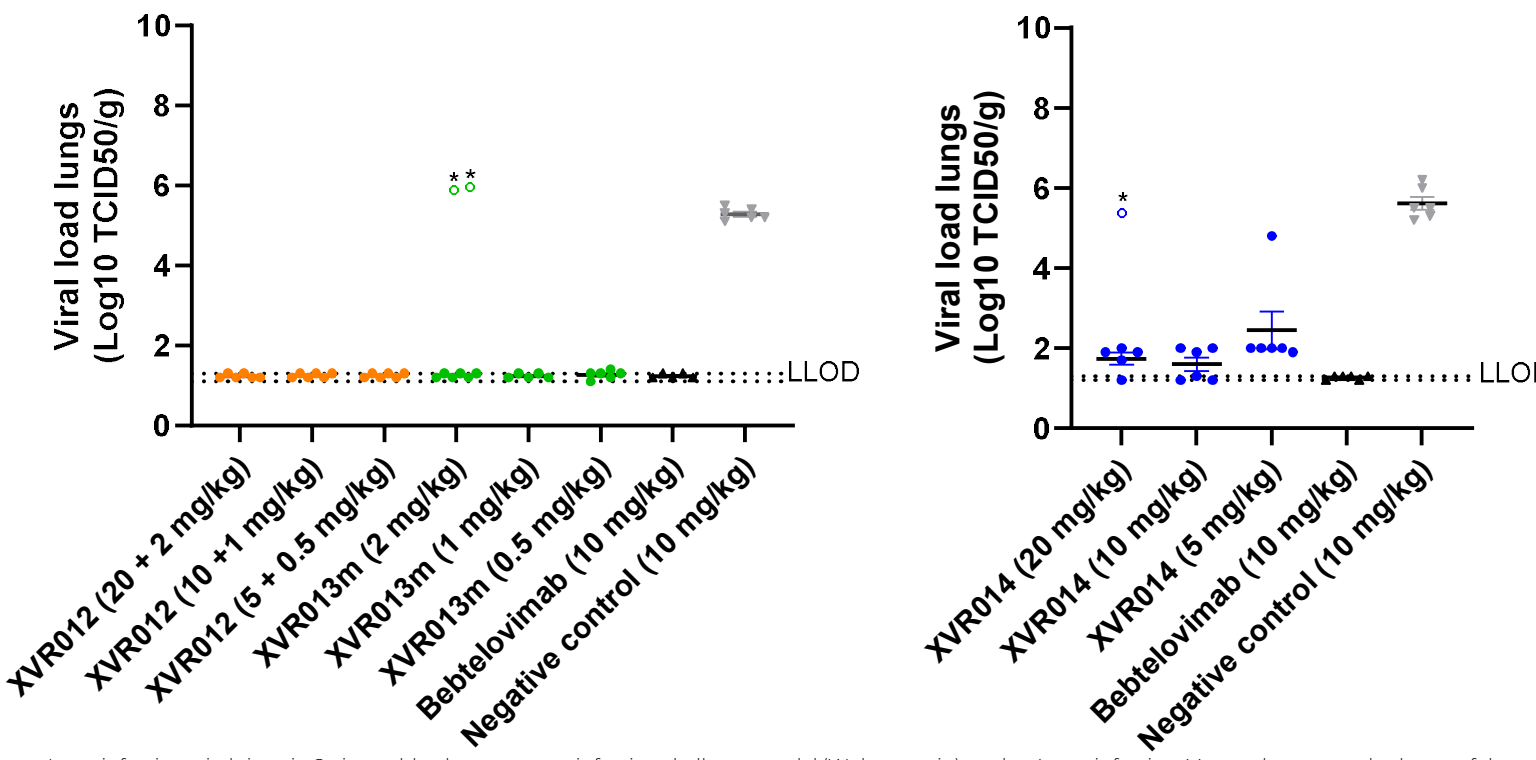
Viral load lungs (Log10 TCID50/g)
0
2
4
6
8
10
LLOD
XVR012 (20 + 2 mg/kg)
XVR012 (10 +1 mg/kg)
XVR012 (5 + 0.5 mg/kg)
XVR013m (2 mg/kg)
XVR013m (1 mg/kg)
XVR013m (0.5 mg/kg)
Bebtelovimab (10 mg/kg)
Negative control (10 mg/kg)
Viral load lungs (Log10 TCID50/g)
LLOD
XVR014 (20 mg/kg)
XVR014 (10 mg/kg)
XVR014 (5 mg/kg)
Bebtelovimab (10 mg/kg)
Negative control (10 mg/kg)

Lung infectious viral titers in Syrian golden hamster post-infection challenge model (Wuhan strain) on day 4 post infection. Mean values + standard error of the mean (SEM) are reported. * Two animals in 2 mg/kg group of XVR013m and one animal in the 20 mg/kg group of XVR014 were experimentally confirmed in PK assays to not have been exposed to the drug. Dotted lines represent the lower limit of detection (LLOD) range.

XVR012 (1:10 ratio of XVR013m:XVR014 at 5,5, 11 and 20 mg/kg), XVR013m (0,5, 1 and 2 mg/kg) and XVR014 (5, 10 and 20 mg/kg) were administered by intraperitoneal injection 4 hours after SARS-CoV-2 challenge (100 TCID50). The negative control group received 10 mg/kg of Palivizumab, a humanized monoclonal antibody directed against the fusion protein of human respiratory syncytial virus. The positive control group received 10 mg/kg of bebtelovimab biosimilar. Six animals were included per group. On day 4 post-infection, animals were euthanized. Right lung lobes were collected and infectious virus was quantified by end-point virus titration. Blood samples were collected before infection and at Day 4 for PK analysis using a qualified ELISA method for the quantitation of XVR012, XVR013m and XVR014 in hamster serum.

- **Infectious viral lung loads** were **completely below detection levels** on Day 4 in all animals experimentally confirmed to have been treated with the high, mid and low doses of both the combination cocktail **XVR012** and **XVR013m**.
- Animals treated with **XVR014** displayed a **dose-dependent effect** on lung viral load. Respectively 1 out of 5 and 3 out of 6 animals in the 20 mg/kg and 10 mg/kg dose groups showed a complete reduction in viral load below detection levels, with the other animals in the group showing an approx. 10.000-fold reduction in infectious viral load compared to animals treated with a negative control antibody. In the 5 mg/kg group, none of the animals showed a full reduction in viral load, but still a 1000 to 10.000-fold reduction in infectious viral load compared to the negative control antibody was observed in all but one animal.

5. Conclusions

- The preclinical data are very promising, demonstrating unchanged virus neutralization potency against all SARS-CoV-2 variants tested in vitro. In vivo, efficacy was demonstrated in the Syrian Golden Hamster model in a therapeutic setting, showing complete reduction of lung viremia for all XVR012 dose levels tested, even with the very low dose of 0,5 mg/kg for the XVR013m component. The allometric scaling based on the PK parameters obtained in an *in vivo* Tg32 SCID mouse model support an envisioned duration of protection up to 6 months in the clinic.
- The 'variant-proof' XVR012 is the first-ever and only combination product that simultaneously targets three highly conserved epitopes, intended for both COVID-19 prevention and treatment. The cocktail XVR012 may provide a long-term solution to the populations at highest risk, including immunocompromised and elderly, for whom there is an urgent unmet medical need.